A comparative analysis of the MSC transcriptome: Human iPSC-derived MSCs and their tissue-derived counterparts

Margeaux Hodgson-Garms[1,2], James Carthew[1], Mikaël Martino[2], Kilian Kelly[3], Jessica E Frith[1,2]

1. Monash University, Department of Materials Science and Engineering, Melbourne Australia. 2. Australian Regenerative Medicine Institute, Monash University, Melbourne Australia. 3. Cynata Therapeutics, Melbourne Australia.

Margeaux.Hodgson-Garms@monash.edu

MargeauxDjaq

FrithLab.com

Background

- Multipotent mesenchymal stromal cells (MSCs) have considerable therapeutic potential and are one of the most popular and versatile cell therapies[1].
- Traditionally sourced from tissue donations, clinical translation is affected by donor-dependence and significant batch-batch, source-based, and intra-population heterogeneity. This limits the predictability and reproducibility of MSCs in the clinic.
- iPSC-derived MSCs (iMSCs) may bypass many of these problems and can potentially provide a limitless supply of batch-consistent, off-the-shelf cell therapies.
- However, how iMSCs compare to tissue-derived MSCs is not yet fully understood.

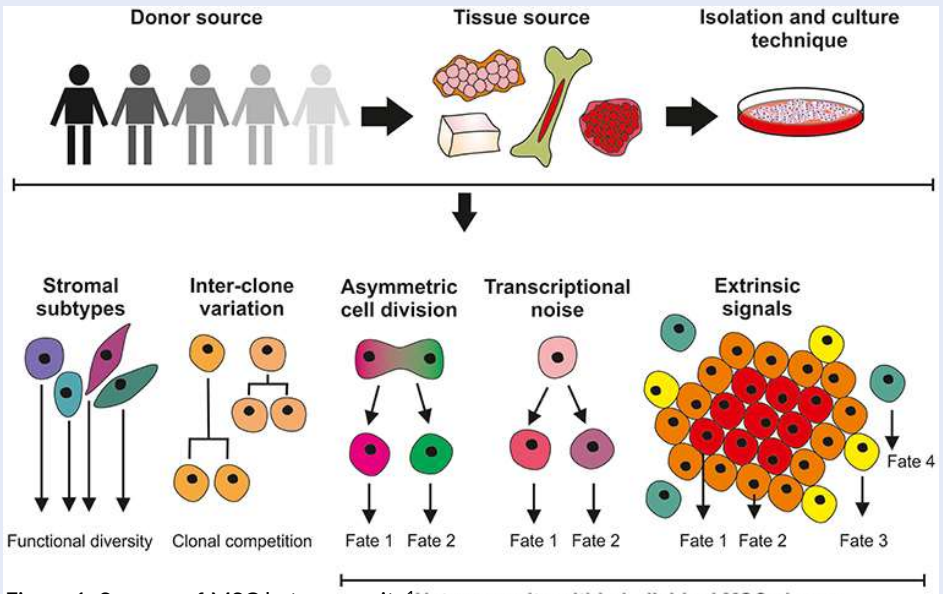

Donor source
Tissue source
Isolation and culture technique
Stromal subtypes
Inter-clone variation
Asymmetric cell division
Transcriptional noise
Extrinsic signals
Functional diversity
Clonal competition
Fate 1
Fate 2
Fate 3
Fate 4
Heterogeneity within individual MSC clones

Figure 1. Sources of MSC heterogeneity[1]

Aims and project design

Aim: To use single cell sequencing to compare and characterise the MSC transcriptome and quantify inter and intra-population intrapopulation heterogeneity between iPSC and tissue-derived MSCs .

This study used NextGen single cell seq to profile transcriptomes of from **13 MSC populations** including multiple batches of clinical grade and commercially available iMSCs, alongside tissue-derived MSCs from bone marrow, adipose tissue and umbilical cord.

After QC and data processing this data set includes transcriptomes from **72,709 individual MSCs** sequenced at a depth of **>100,000 reads/ cell.**

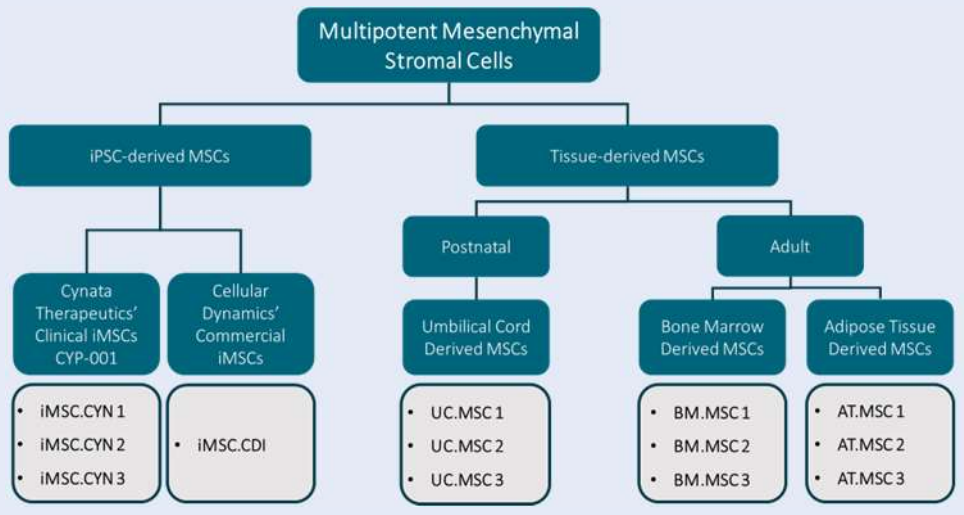

Multipotent Mesenchymal Stromal Cells
iPSC-derived MSCs
Tissue-derived MSCs
Postnatal
Adult
Cynata Therapeutics' Clinical iMSCs CYP-001
Cellular Dynamics' Commercial iMSCs
Umbilical Cord Derived MSCs
Bone Marrow Derived MSCs
Adipose Tissue Derived MSCs
iMSC.CYN 1
iMSC.CYN 2
iMSC.CYN 3
iMSC.CDI
UC.MSC 1
UC.MSC 2
UC.MSC 3
BM.MSC 1
BM.MSC 2
BM.MSC 3
AT.MSC 1
AT.MSC 2
AT.MSC 3

Figure 2. Schematic outline of the source and labelling of MSC populations used.

MSCs cluster primarily by tissue/ source.

UMAP clustering of MSC transcriptomes indicates that tissue/ source of origin accounts for most MSC heterogeneity **(Fig.3A)**. MSC tissue/ sources formed clades within themselves. BM.MSC and AT.MSCs branched latest while iMSC and UC.MSCs branched earlier indicating comparatively less similarity **(Fig.3B).**

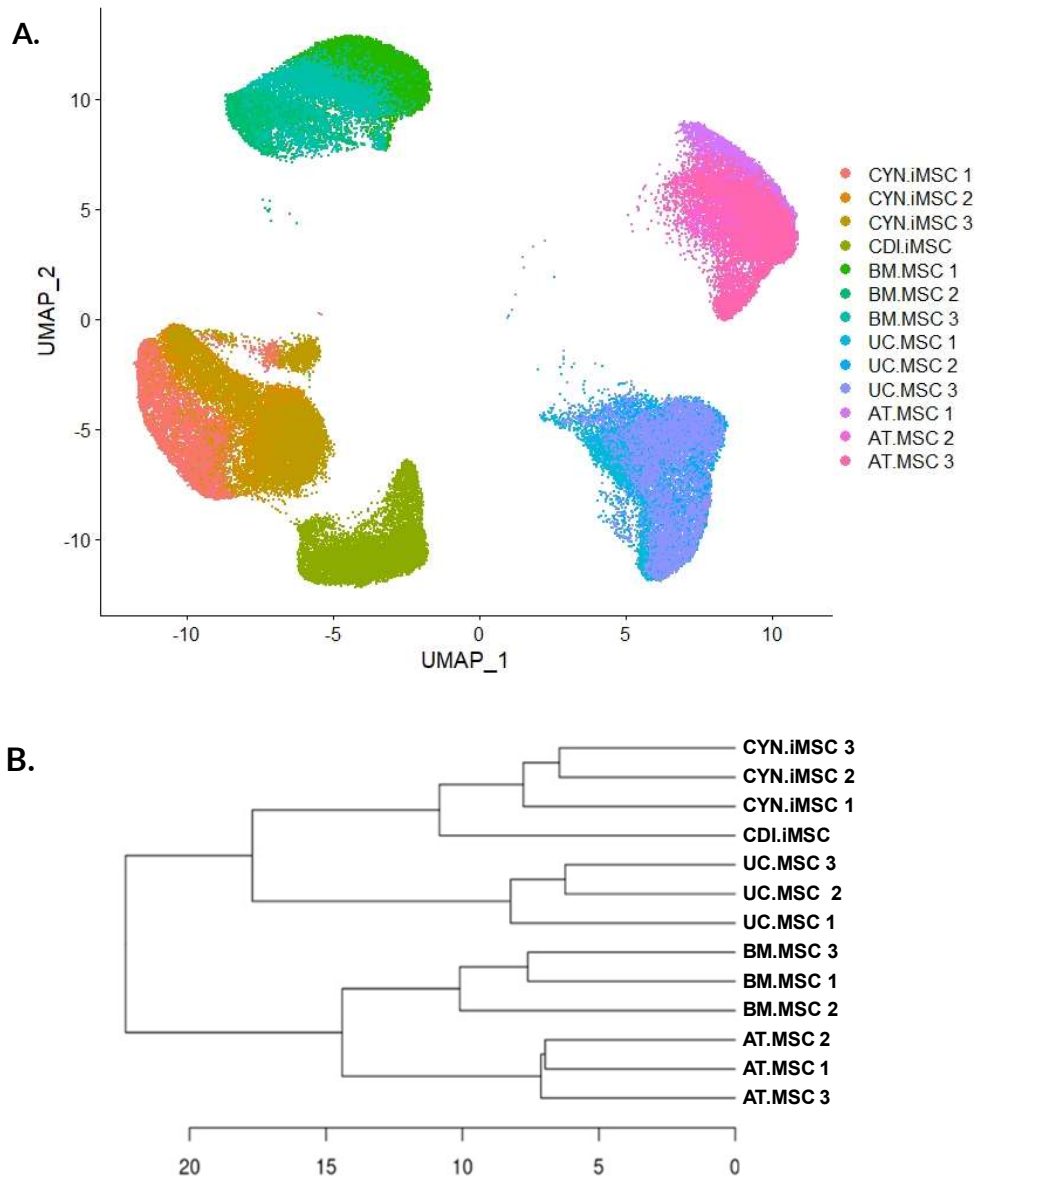

A.
UMAP_2
UMAP_1
CYN.iMSC 1
CYN.iMSC 2
CYN.iMSC 3
CDI.iMSC
BM.MSC 1
BM.MSC 2
BM.MSC 3
UC.MSC 1
UC.MSC 2
UC.MSC 3
AT.MSC 1
AT.MSC 2
AT.MSC 3
B.
CYN.iMSC 3
CYN.iMSC 2
CYN.iMSC 1
CDI.iMSC
UC.MSC 3
UC.MSC 2
UC.MSC 1
BM.MSC 3
BM.MSC 1
BM.MSC 2
AT.MSC 2
AT.MSC 1
AT.MSC 3

Figure 3. UMAP visualisation of MSC transcriptome clustering (A). Single-cell MSC transcriptomes (N=72,809) from 13 populations (indicated by point colour) visualised using UMAP projection generated in Seurat using function DimPlot. **Dendrogram of MSC population hierarchy (B).** Seurat hclust function was used to generate a hierarchical population (n=13) dendrogram based on single-cell distance-from centroid-values. Branching depth (X) indicates population similarity.

Differentially expressed (DE) genes were identified between iMSC and tissue-derived MSCs.

820 genes were upregulated in tissue-derived MSCs (tMSCs) while 5491 genes were upregulated in iMSCs **(Fig. 4A)**. Gene Ontology (GO) term enrichment analysis was used to query DE genes for enriched Biological Processes (BP). BP including telomere maintenance and RNA catabolism processes were enriched in genes upregulated in iMSCs, while genes downregulated in iMSCs were enriched for humoral immune response and complement processes **(Fig. 4B)**.

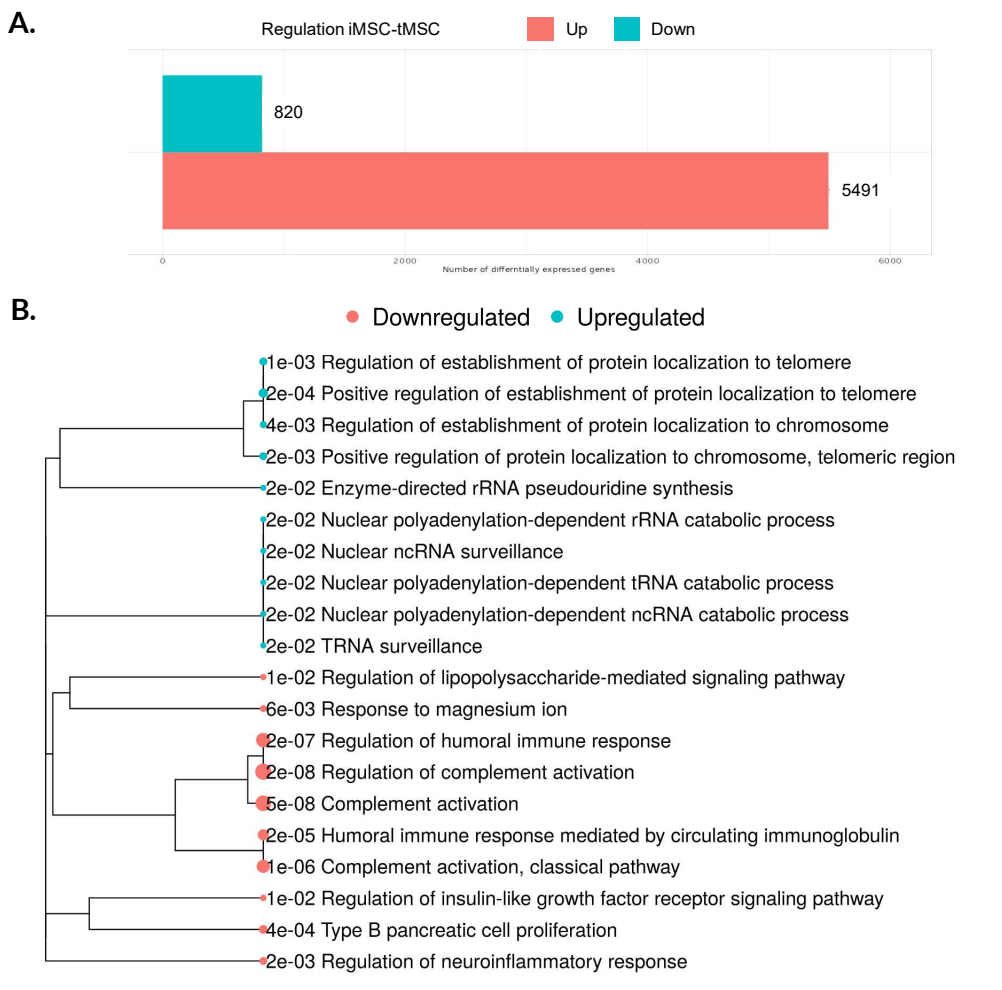

A.
Regulation iMSC-tMSC
Up
Down
820
5491
Number of differentially expressed genes
B.
Downregulated
Upregulated
1e-03 Regulation of establishment of protein localization to telomere
2e-04 Positive regulation of establishment of protein localization to telomere
4e-03 Regulation of establishment of protein localization to chromosome
2e-03 Positive regulation of protein localization to chromosome, telomeric region
2e-02 Enzyme-directed rRNA pseudouridine synthesis
2e-02 Nuclear polyadenylation-dependent rRNA catabolic process
2e-02 Nuclear ncRNA surveillance
2e-02 Nuclear polyadenylation-dependent tRNA catabolic process
2e-02 Nuclear polyadenylation-dependent ncRNA catabolic process
2e-02 TRNA surveillance
1e-02 Regulation of lipopolysaccharide-mediated signaling pathway
6e-03 Response to magnesium ion
2e-07 Regulation of humoral immune response
2e-08 Regulation of complement activation
5e-08 Complement activation
2e-05 Humoral immune response mediated by circulating immunoglobulin
1e-06 Complement activation, classical pathway
1e-02 Regulation of insulin-like growth factor receptor signaling pathway
4e-04 Type B pancreatic cell proliferation
2e-03 Regulation of neuroinflammatory response

Figure 4. Differential expression iMSC vs tMSCs (A). DeSeq2 was used to identify genes significantly up (N=5491) or downregulated (N-820) between iMSC and tMSCs. **Top biological processes enriched in DE genes (B).** GO term enrichment analysis was used to identify the top 10 most strongly enriched BP both upregulated and downregulated in iMSCs. GO term tree was generated based on shared gene membership. Point size is representative of enrichment score and point colour indicates if gene members are up or down regulated. Plots were generated in iDEP 1.0 [4]

Intrapopulation variance was quantified as a factor of cell-cell gene variance within the top 200 most variable genes.

Mean cell-cell transcriptomic variance was observed to be significantly lower in iMSCs than tMSCs. Furthermore, mean cell-cell variance was comparable between iMSC populations while tMSC populations showed significant donor-donor differences.

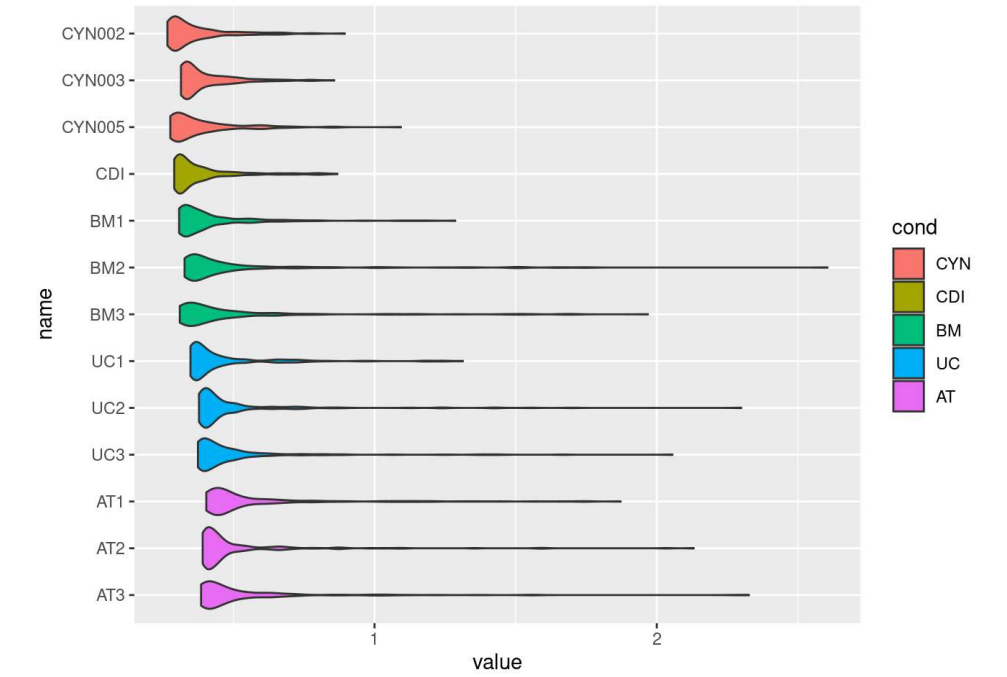

CYN002
CYN003
CYN005
CDI
BM1
BM2
BM3
UC1
UC2
UC3
AT1
AT2
AT3
name
value
cond
CYN
CDI
BM
UC
AT

Figure 6. Violin plots of cell-cell variance across top 200 most variable genes. The top 200 most variable genes were identified using DeSeq2. Gene-wise variance from the median was calculated for each cell and single-cell. Variance scores (x) are presented as a violin plot. Tissue/ source is indicated by colour. Plot was produced using Seurat.

Principle Component Analysis (PCA) and PCA loading identify gene markers driving separation of iMSCs.

PCA of MSC transcriptomes identified two components accounting for 40.47% and 20.16% of variation between MSC tissue/sources. PCA loading identified major genes driving component separation **(Fig 5A)**. Expression of CRMP1, DIPK1B and LIN28B was found to drive separation of iMSCs from tMSCs, with highest expression in iMSCs followed by BM.MSCs, UC.MSC and AT.MSCs **(Fig 5B).**

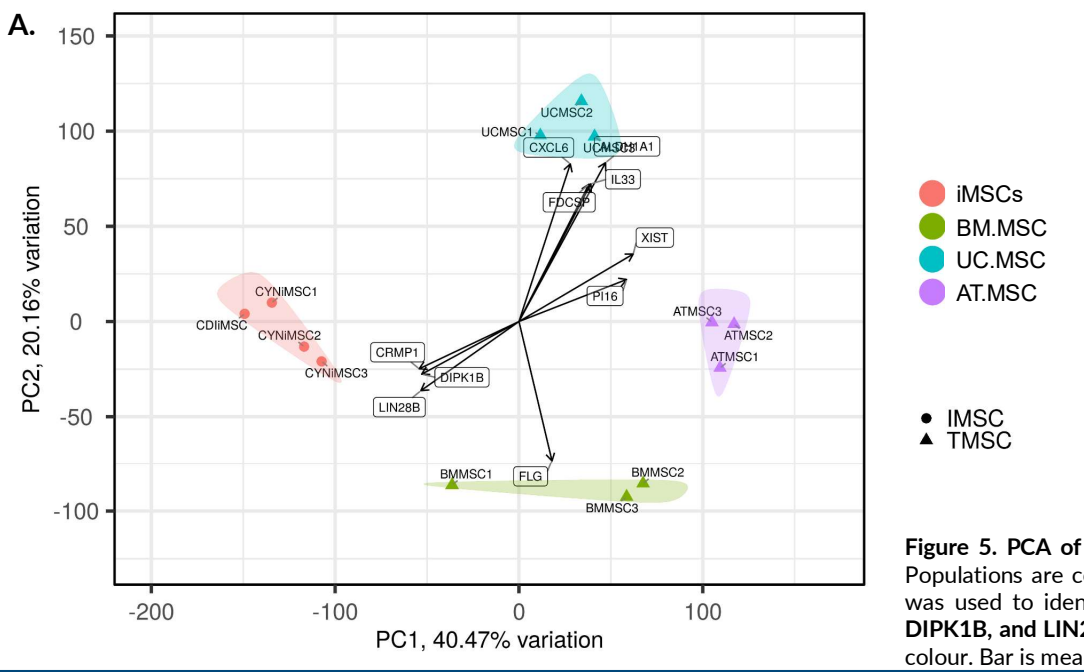

A.
PC2, 20.16% variation
PC1, 40.47% variation
CRMP1
DIPK1B
LIN28B
CXCL6
IL33
FDCSP
XIST
PI16
FLG
iMSCs
BM.MSC
UC.MSC
AT.MSC
IMSC
TMSC

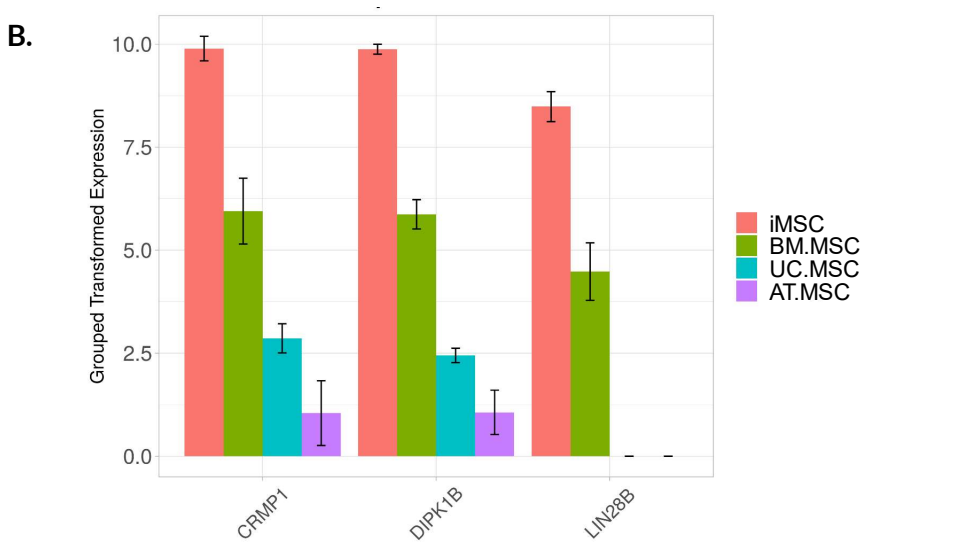

B.
Grouped Transformed Expression
CRMP1
DIPK1B
LIN28B
iMSC
BM.MSC
UC.MSC
AT.MSC

Figure 5. PCA of MSC populations (A). PCA was used to visualise components of MSC tissue/ source separation. Populations are coloured by tissue/ source and marker shape indicates iMSC vs tMSC grouping. PCAtools Package was used to identify loading genes driving iMSC/ tMSC separation. **Expression of major loading genes CRMP1, DIPK1B, and LIN28B (B).** Expression of loading genes is presented as bar plots with MSC tissue/ source indicated by colour. Bar is mean grouped transformed expression ± SD.

Conclusions

Key Findings:

1) Tissue/ source is the primary driver of MSC heterogeneity.
2) iMSCs are most closely related to UC.MSCs, while BM.MSCs and AT.MSCs are more closely related to each other.
3) iMSCs differ from tissue-derived MSCs by the upregulation of biological processes linked to telomere maintenance and RNA catabolism, and the downregulation of humoral immune response and complement processes.
4) iMSCs exhibit less batch-batch heterogeneity than tissue-derived MSCs, furthermore they also exhibit significantly less intra-population variation.

This data set provides a comprehensive profile of MSC transcriptomes at a single-cell level, allowing us to develop a better understanding of the sources of MSC heterogeneity and improve predictability of clinical outcomes. Moreover, this study confirms that iMSCs successfully bypass much of the inherent heterogeneity that affects the clinical application of tissue-derived MSCs, validating their promise as an off-the-shelf cell therapy.

References and Acknowledgments

1. ClinicalTrials.gov. Search of: Mesenchymal Stem Cell - List Results - ClinicalTrials.gov. https://clinicaltrials.gov/ct2/results?cond=&term=Mesenchymal+Stem+Cell&cntry=&state=&city=&dist=.
2. Wilson, A., Hodgson-Garms, M., Frith, J. E. & Genever, P. Multiplicity of mesenchymal stromal cells: Finding the right route to therapy. *Front. Immunol.* **10**, (2019).
3. Dominici, M. et al. 2006. Minimal criteria for defining multipotent mesenchymal stromal cells. The International Society for Cellular Therapy position statement. *Cytotherapy*, 8(4), pp.315-317.
4. Ge, S. X., Son, E. W. & Yao, R. iDEP: an integrated web application for differential expression and pathway analysis of RNA-Seq data. *BMC Bioinforma. 2018 191* **19**, 1–24 (2018).

This project was supported by:
Australian Govt. RTP Stipend
Monash University Dpt. of Materials Science and Engineering
Monash University Graduate Research Completion Award